ANAVEX 7-1037, A NOVEL SMALL SYNTHETIC SIGMA LIGAND, INDUCES APOPTOSIS IN HUMAN HCT116 COLON CANCER CELLS *IN VITRO* AND SUPPRESSION OF HCT116 TUMOR GROWTH IN SCID MICE.

Mahaira G L [2], H Khan 2, C Tsimblouli[1], P Pantazis [1], E Karamitopoulou [2], A Vamvakides [2] and K Dimas [1]

1;Laboratory of Pharmacology-Pharmacotechnology, Biomedical Research Foundation of the Academy of Athens, 4 Soranou Efessiou Str, Athens, Greece,
2;Eurogenet Labs SA, Marathonos Av. 27, Athens, Greece

- Sigma receptors are unique drug-binding proteins present in the central nervous system and in various peripheral tissues.
- It has been shown that sigma receptor subtypes are highly expressed in tumor cell lines from various tissues.
- Their high density in tumor and proliferating cells, makes sigma receptors potential target for diagnostic imaging as well as therapeutic agents.
- In the present study the compound Anavex 7-1037 **(C7**) was tested [chlorohydrate of 5-(Tricyclo[3,3,1,13,7]dec-1-yl)-dihydro-3-(dimethylamino-methyl)-5-phenylfuran-2(3H)-one].
- **C7** is a low molecular weight, synthetic compound exhibiting high affinity for sigma-1 (nanomolar) and moderate (micromolar) affinity for sigma-2 and sodium channels.

In vitro studies

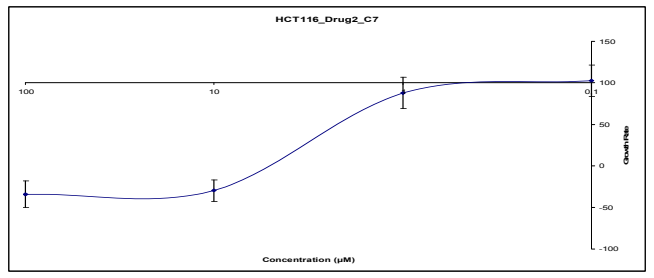
HCT116_Drug2_C7
100
10
0.1
150
100
50
0
-50
-100
Growth Rate
Concentration (µM)

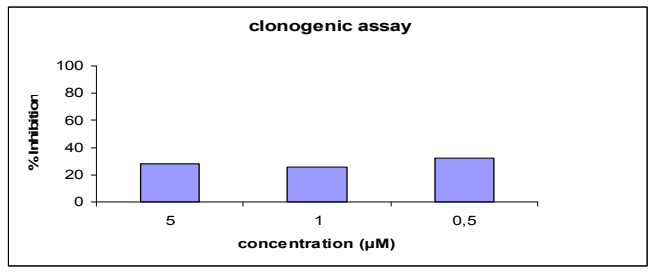
clonogenic assay
100
80
60
40
20
0
% Inhibition
5
1
0,5
concentration (µM)

SHORT TERM SRB ASSAY (2-days 5-dose) :
activity at low micromolar range.

LONG TERM CLONOGENIC ASSAY (14-days 3-dose):
activity below 1μM.

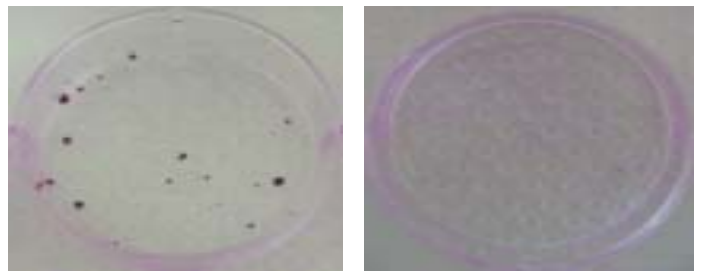

CNT **C7**

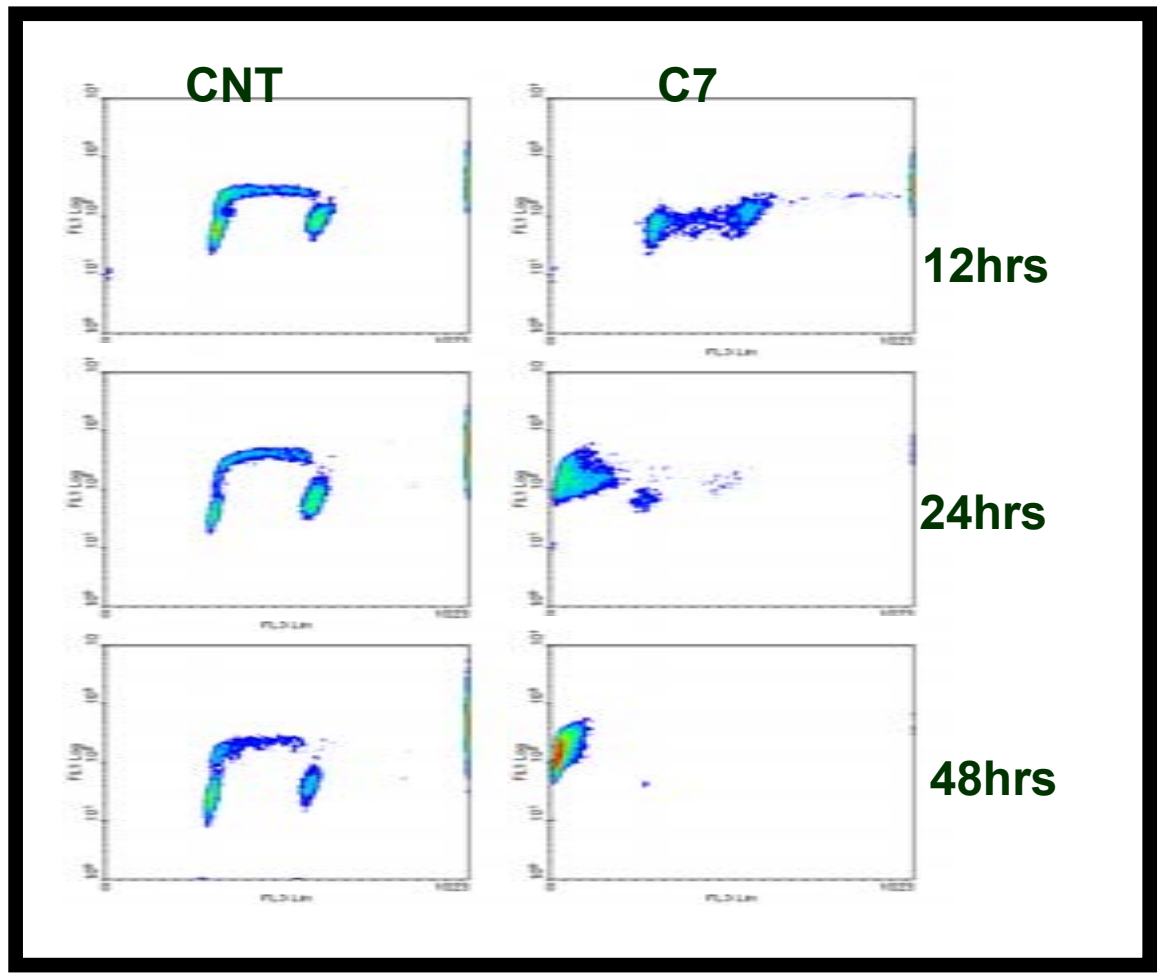
CNT
C7
12hrs
24hrs
48hrs

C7-EFFECT ON CELL CYCLE

HCT116 cells were treated with **C7** for the indicated time periods. Cells were labeled with BrdUdr, harvested and subjected to flow cytometry analysis, demonstrating arrest of the cell cycle.

MECHANISM OF CELL DEATH

C7 induces detachment of cells and apoptosis via activation of caspase 3, caspases -8 and -9 and PARP as was estimated by western blot analysis.

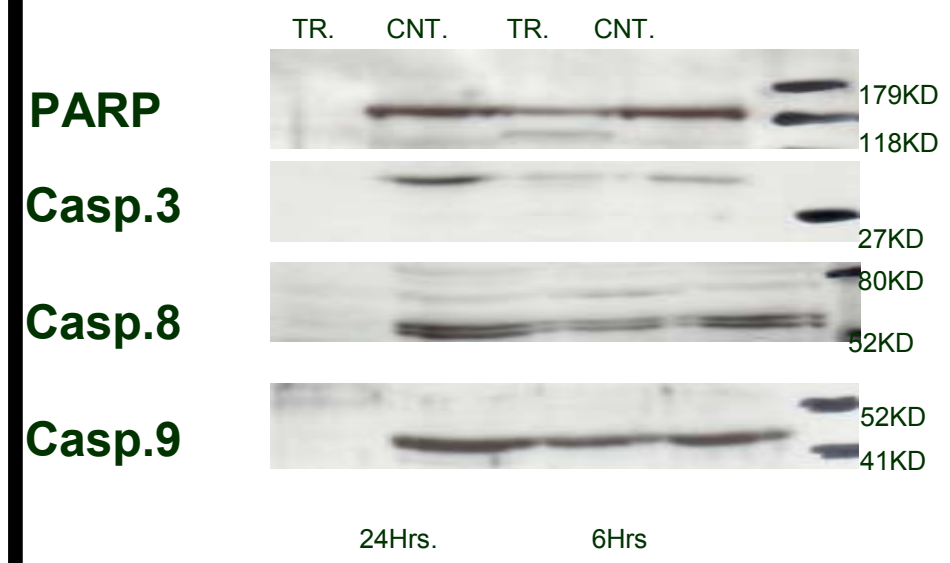
TR.
CNT.
TR.
CNT.
PARP
Casp.3
Casp.8
Casp.9
179KD
118KD
27KD
80KD
52KD
52KD
41KD
24Hrs.
6Hrs

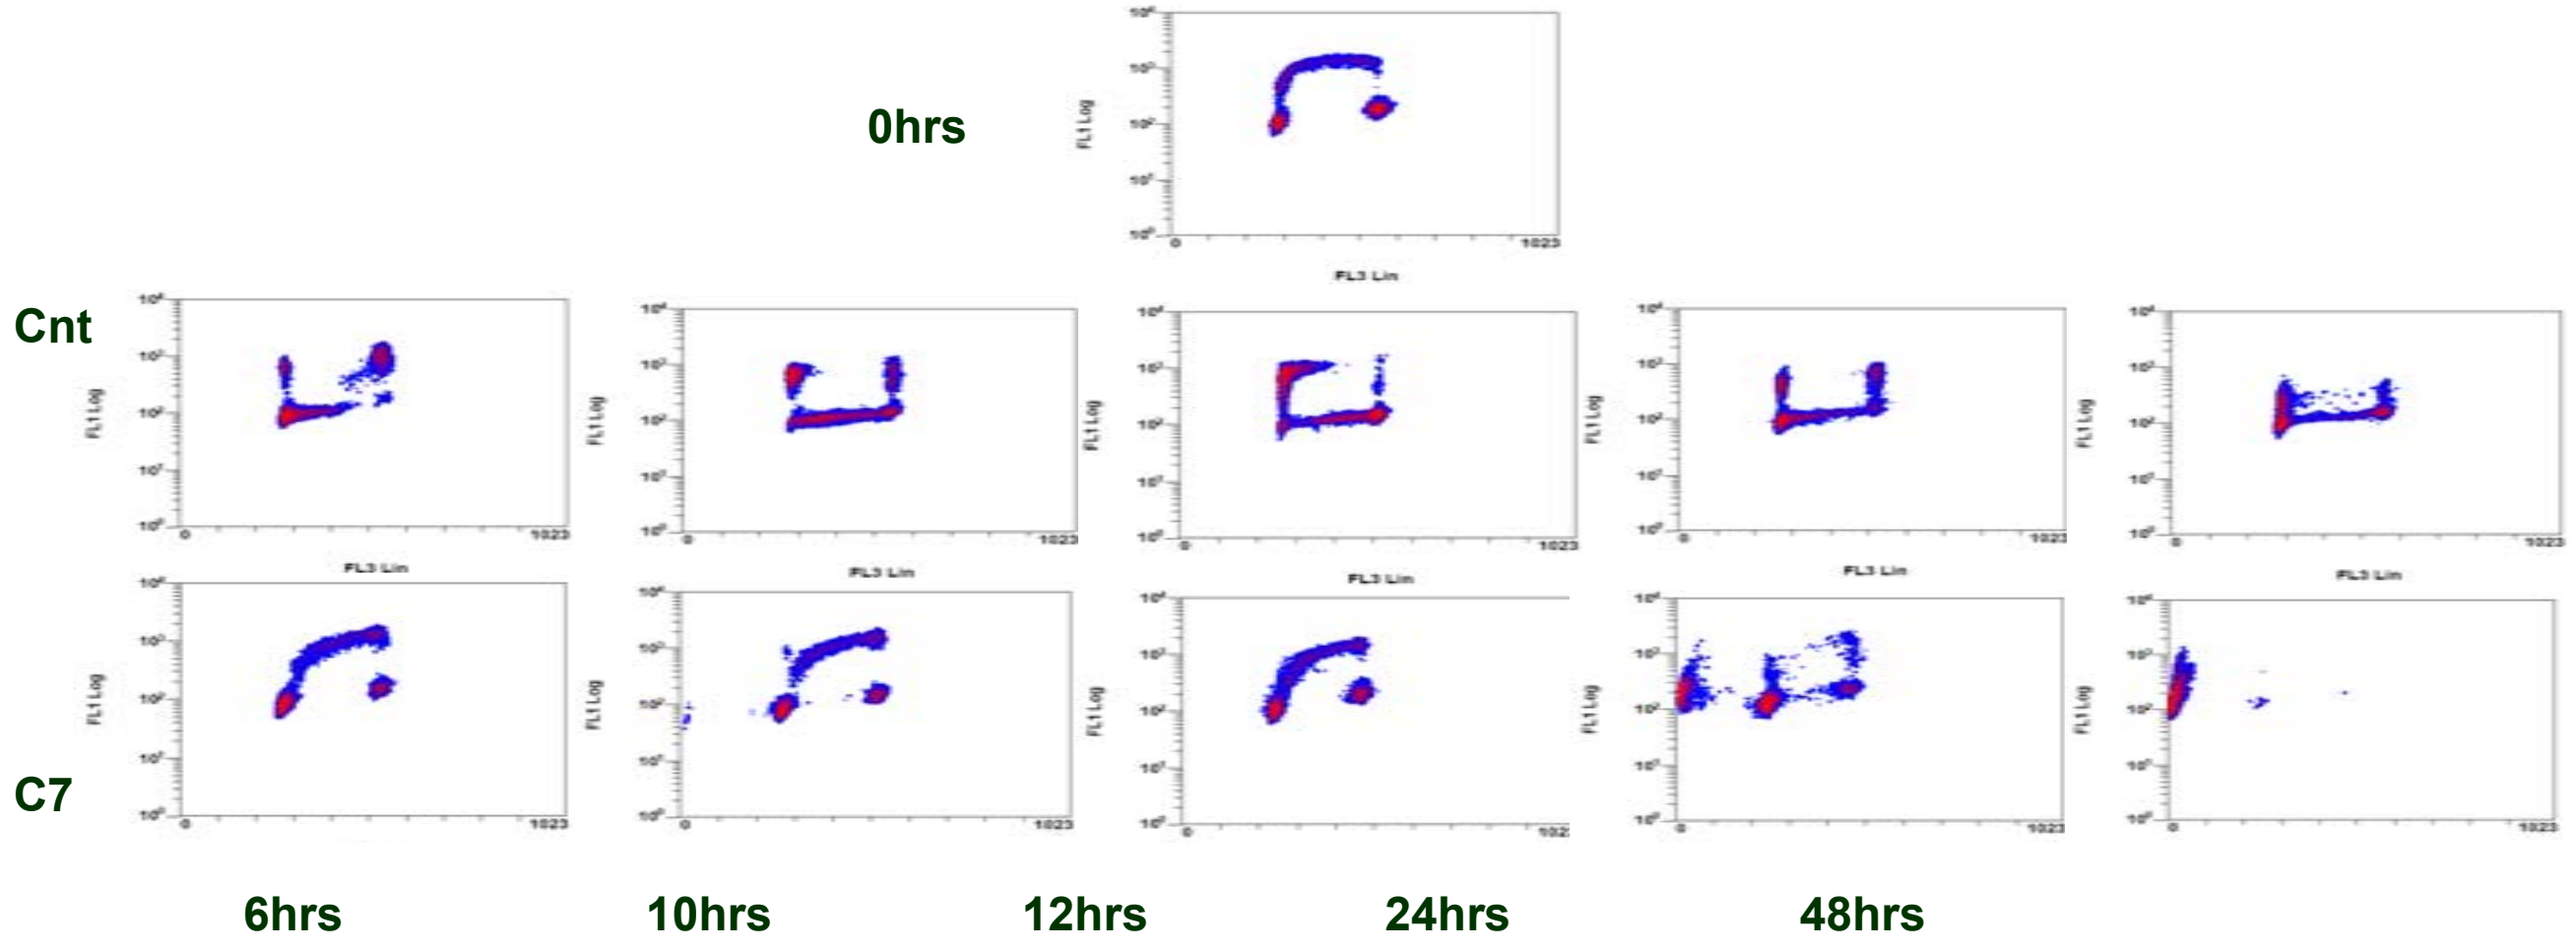
0hrs
Cnt
C7
6hrs
10hrs
12hrs
24hrs
48hrs
FL1 Log
FL3 Lin

C7-INDUCED APOPTOTIC POPULATION (DNA KINETICS).

HCT116 cells were initially labeled with BrdUdr and then **C7** was added for the indicated periods of time. Cells were harvested and subjected to flow cytometry analysis indicating that the apoptotic population derives mainly from the S-phase.

In vivo studies

C7-TREATMENT OF HCT116-XENOGRAFTED SCID MICE

C7 was administered i.p. following the Q4D3 schedule repeated for two weeks.
Average tumor size (in mm^3) was calculated for untreated mice(•) and treated (100mg/kg,■).

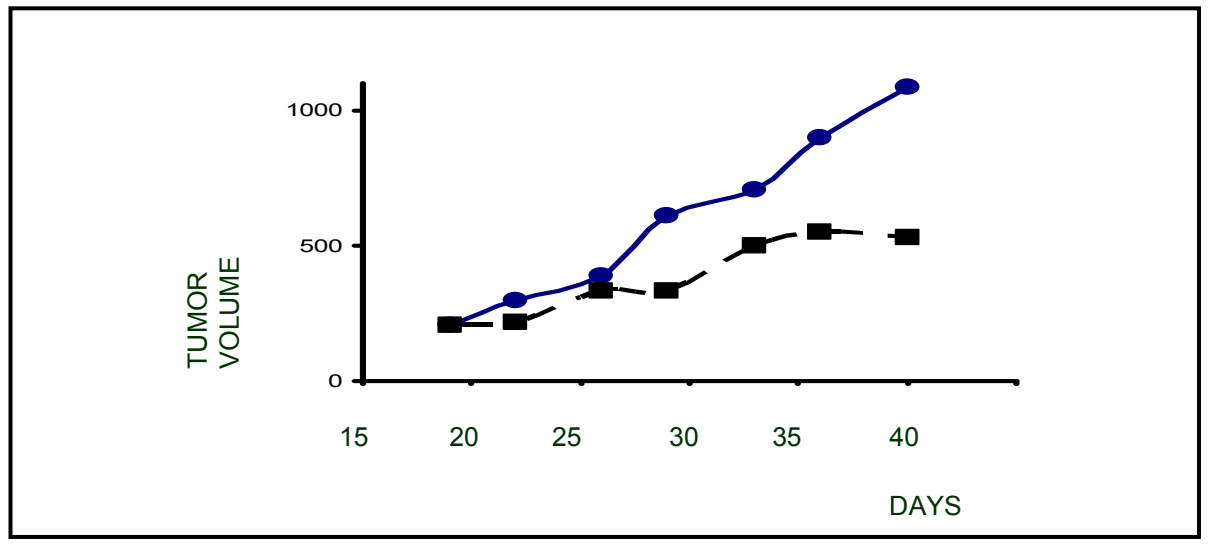
1000
500
0
TUMOR VOLUME
15
20
25
30
35
40
DAYS

Conclusions

ANAVEX 7-1037 (C7)

- exhibits significant GI50 potency
- induces arrest and apoptosis affecting initially those cells that are in the S-phase of the cycle
- can significantly delay the growth of HCT116 xenografts
- This and other patented original sigma ligands are currently tested in vivo on different xenografts (prostate, breast, melanoma etc).